Laminar Wash™ AUTO system: a reliable walk-away sample preparation solution for better TILs recovery without centrifugation

Kim I[1], Lye M[1], Zappasodi R[2], Schulze I[2], Eberle C[3], Ke CY[1], Fernandez Oropeza N[1], Wang A[1], Cheng KL[1], Kon IC [1], Pek R[1], Merghoub T[2], Kim N[1]
Curiox Biosystems Inc., MA, USA[1], Memorial Sloan Kettering Cancer Center, NY, USA[2], Charles River Laboratories, MA, USA [3]

CURIOX'S LAMINAR WASH TECHNOLOGY AND FULL AUTOMATION OF SAMPLE PREPARATION WITH LW™ AUTO1000

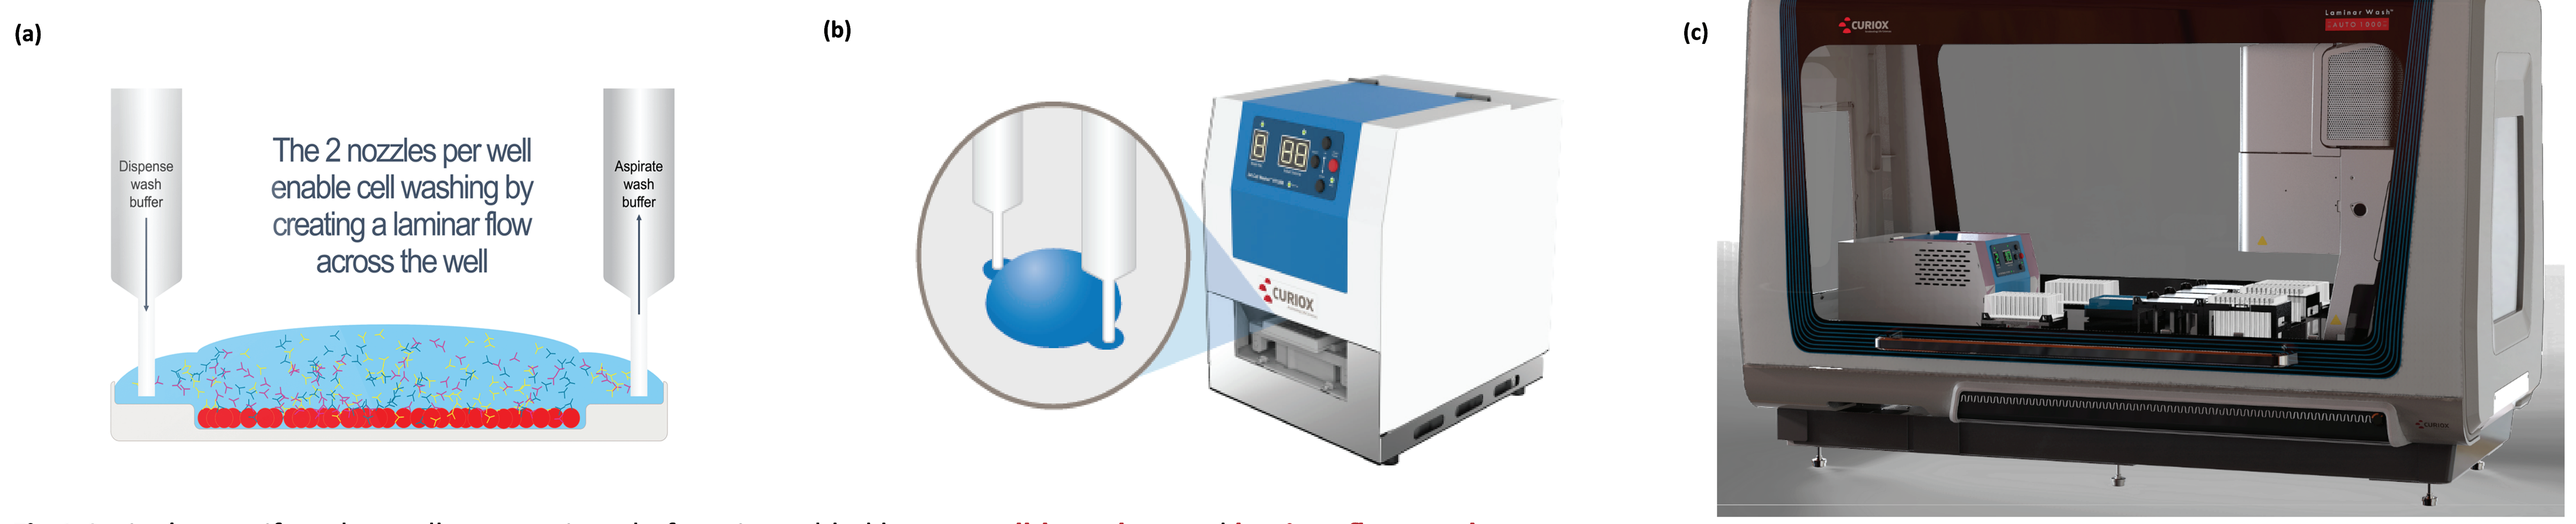
(a)
Dispense wash buffer
The 2 nozzles per well enable cell washing by creating a laminar flow across the well
Aspirate wash buffer
(b)
(c)

Fig 1 Curiox's centrifuge-less cell preparation platform is enabled by our **wall-less plate** and **laminar flow washer**.
(a) The Laminar Wash™ (LW) plate (96 well) or strip (16 well) consists of an array of hydrophilic spots surrounded by hydrophobic surface, which functions as a virtual wall that separates each spot. Each spot can process from a single cell to as many as 10 million cells without the mechanical stress and cell losses associated with centrifugation.
(b) HT1000 washes 96 wells in a fully automated process with individual nozzles for each well.
(c) The **Laminar Wash™ AUTO 1000** software interface allows modifications to sample volume, buffer volume, antibody volume, number of washes, incubation time and temperature. In addition, it comes with pre-programmed protocols to ensure user consistency. **User-friendly Graphical Interface (GUI):** The software prompts the user to conveniently enter the parameters of existing SOPs or protocols with no coding or programming of the liquid handling system.

AUTOMATED LAMINAR WASH™ PRODUCES CONSISTENT DATA with LOW COEFFICIENT of VARIANCE

(a)

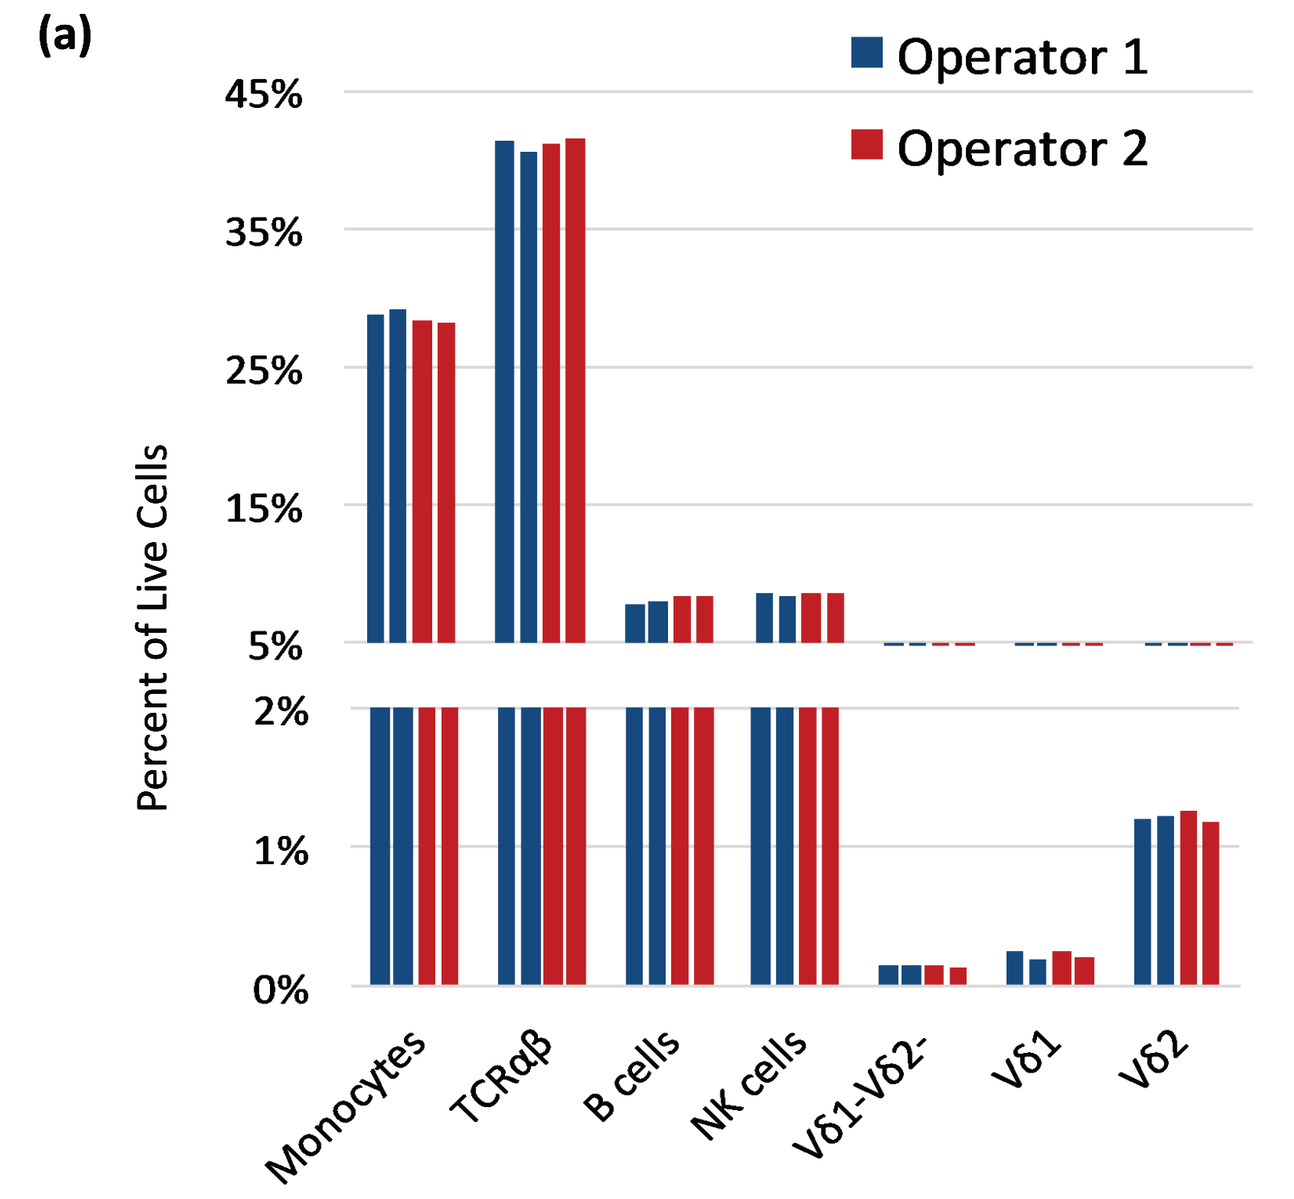
Operator 1
Operator 2
Percent of Live Cells
45%
35%
25%
15%
5%
2%
1%
0%
Monocytes
TCRαβ
B cells
NK cells
Vδ1-Vδ2-
Vδ1
Vδ2

(b)

Population	% CV
Monocytes	1.5
TCRαβ	1.0
B cells	3.4
NK cells	1.2
Vδ1	11.1
Vδ1-Vδ2-	10.1
Vδ2	2.4

Conventional method ~20% CV

Fig 2 By using Laminar Wash™ in a single wash step during immunophenotyping of human PBMCs, the user achieved **(a)** consistency and **(b)** lower coefficient of variance even between different operators. *Webinar available from www.curiox.com*

LW™ BETTER REMOVES STAINING BACKGROUND LEADING TO TRUE POSITIVE SIGNAL DETECTION

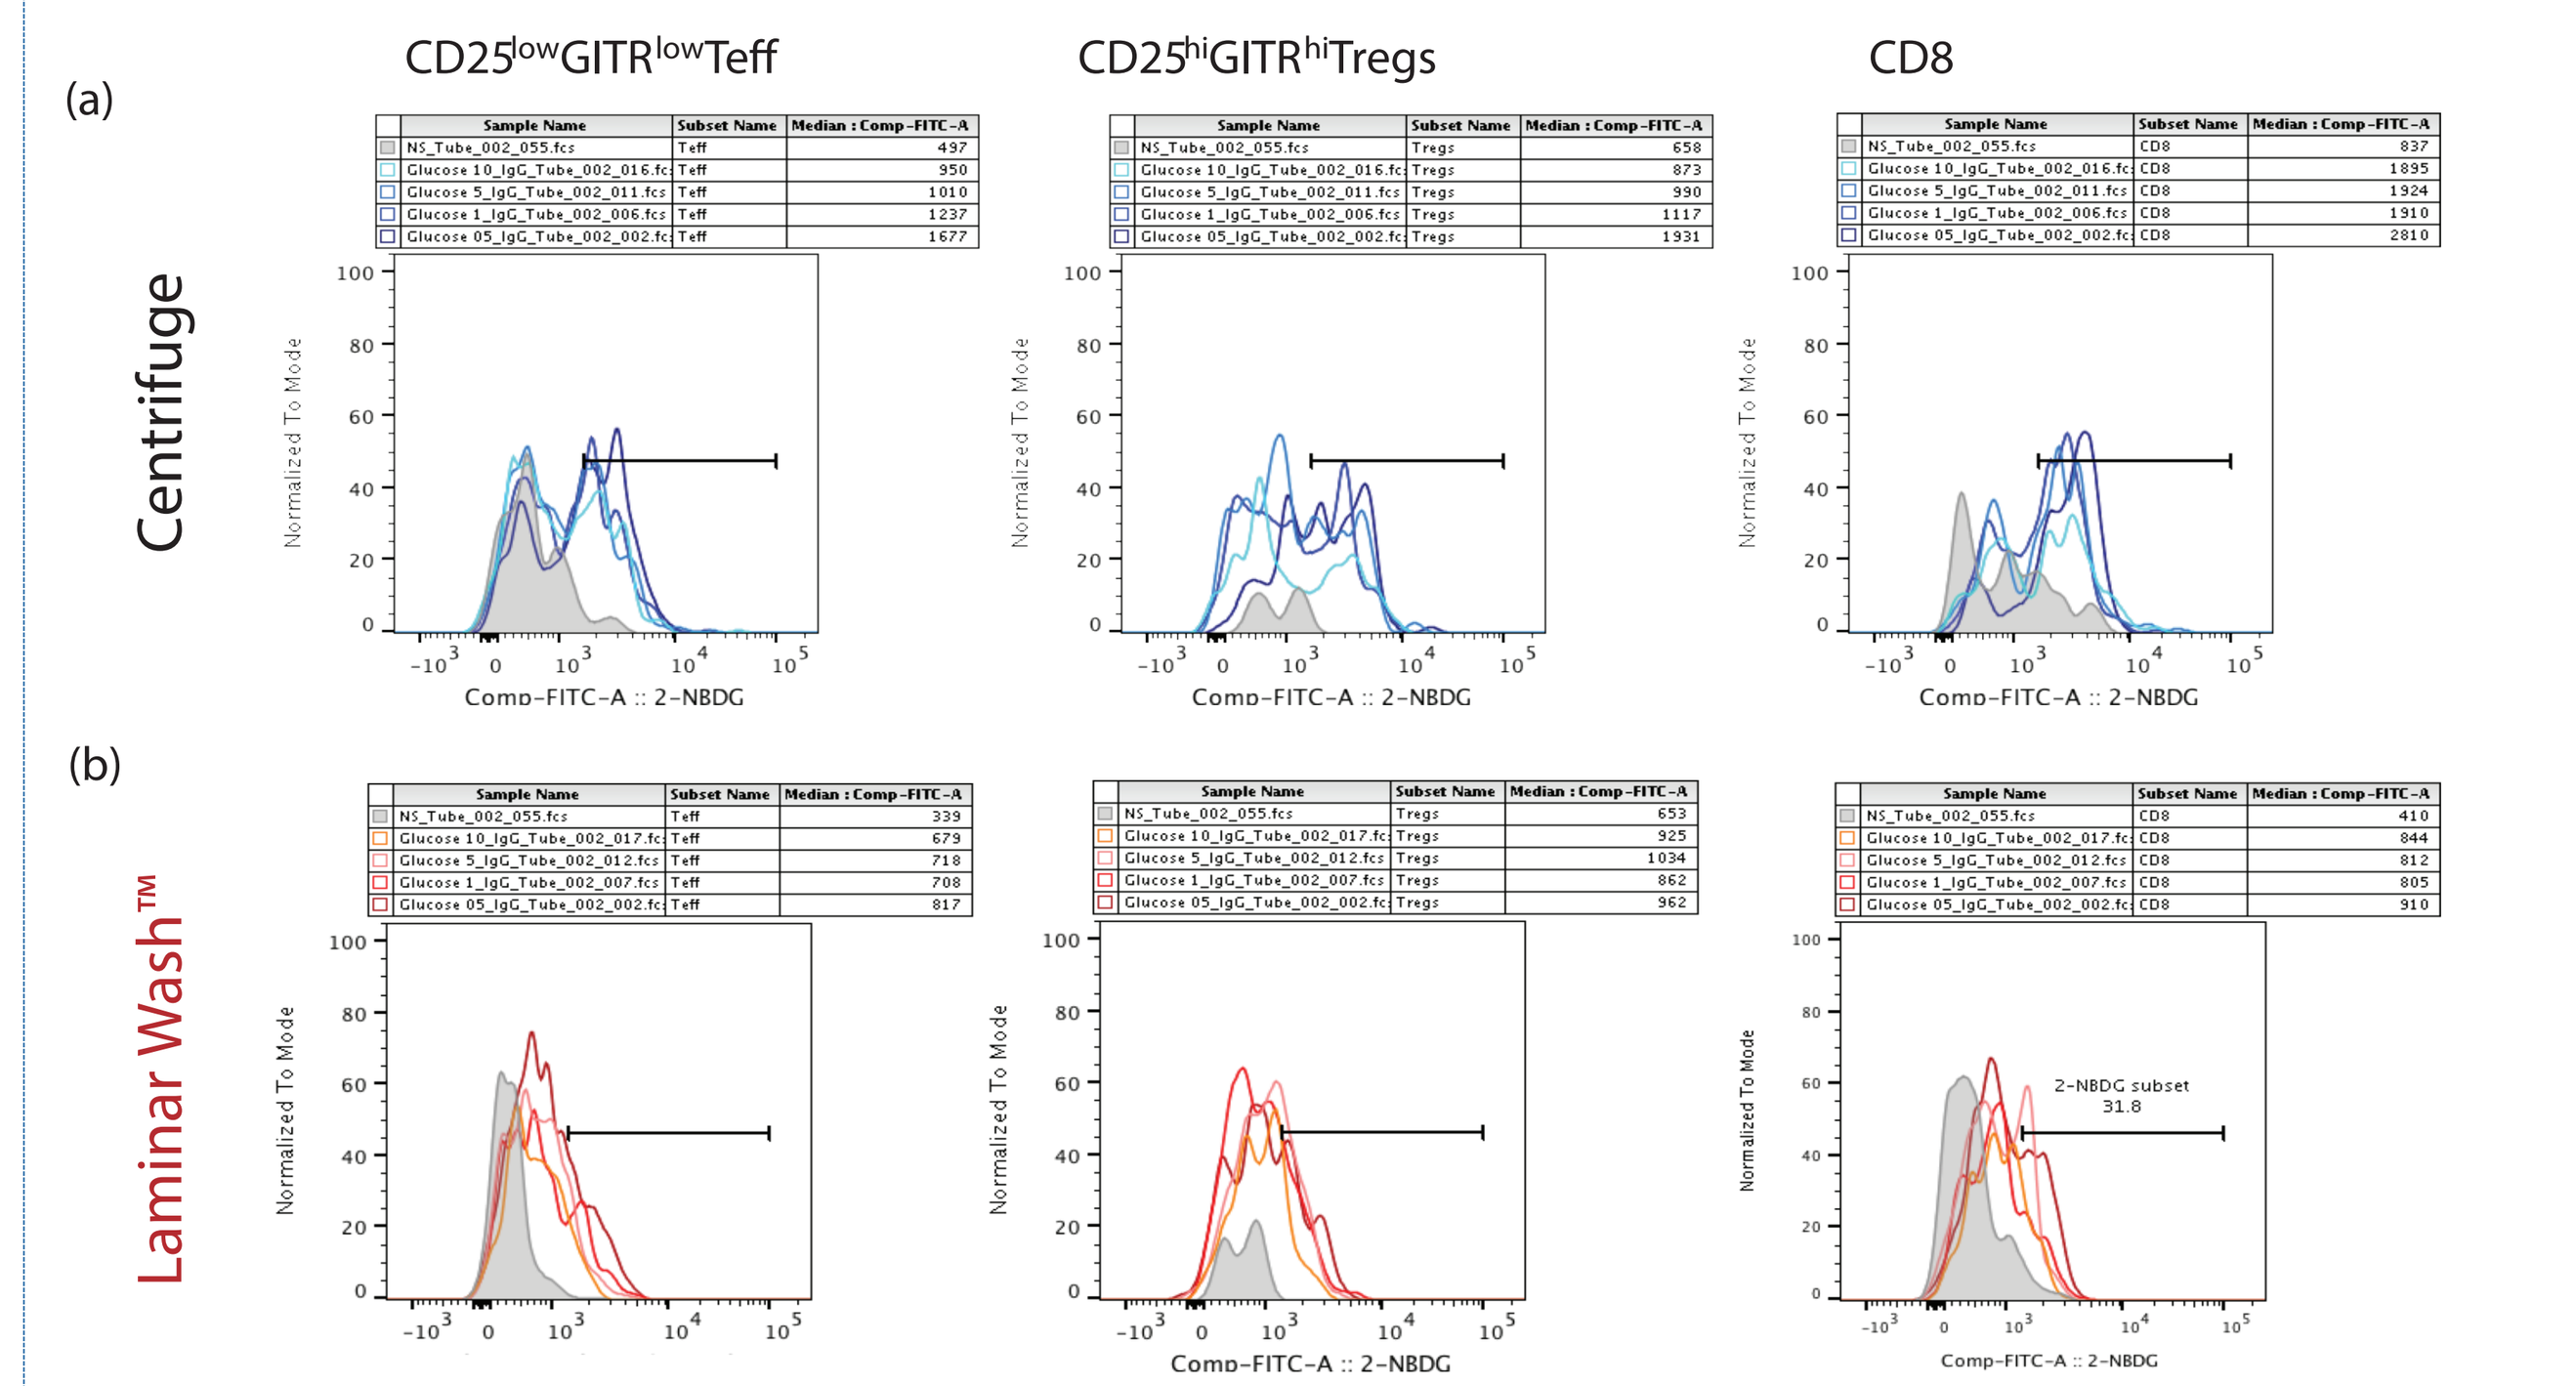
(a)
(b)
CD25lowGITRlowTeff
CD25hiGITRhiTregs
CD8
Centrifuge
Laminar Wash™
Comp-FITC-A :: 2-NBDG
Normalized To Mode
2-NBDG subset 31.8

Fig 6 Splenocytes from 4T1 bearing BALB/c activated for 24hrs with soluble aCD3 (0.5µg/ml) in the presence of glucose [0.5 10 mM]. Cultures were then washed and incubated with 2-NBDG (final concentration 100mM). Cells were washed by (a) Centrifuge vs. (b) Laminar Wash method.
*NS= non stimulated, but fully stained

GATING STRATEGIES for the 2-NBDG ASSAY

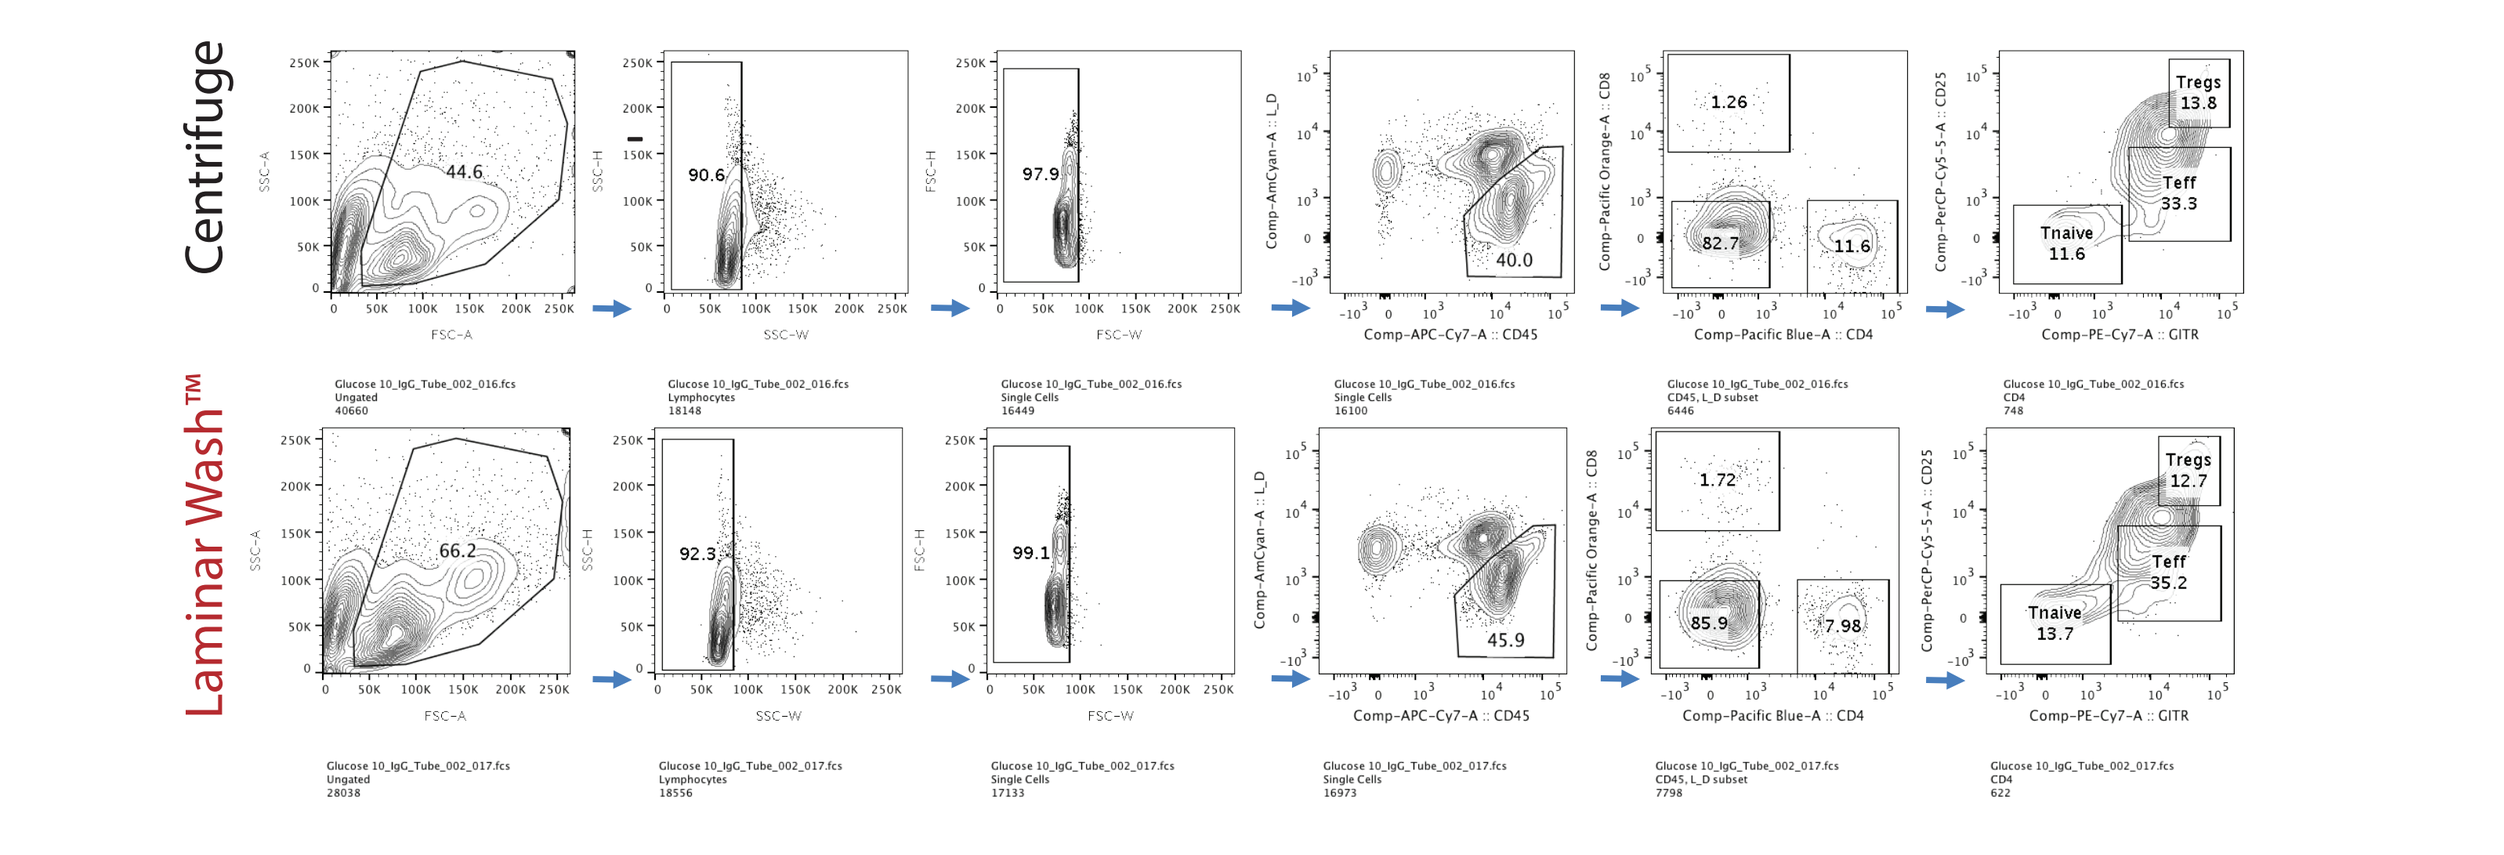
Centrifuge
Laminar Wash™
44.6
90.6
97.9
40.0
1.26
82.7
11.6
Tregs 13.8
Teff 33.3
Tnaive 11.6
66.2
92.3
99.1
45.9
1.72
85.9
7.98
Tregs 12.7
Teff 35.2
Tnaive 13.7

- After extensive washing, cells were stained with a panel of fluorochrome conjugated antibodies for surface markers to identify CD8 and CD4 effector T cells as well as Tregs

LAMINAR WASH™ IMPROVES RETENTION and BETTER RETAINS ENDOGENOUS PROTEIN EXPRESSION

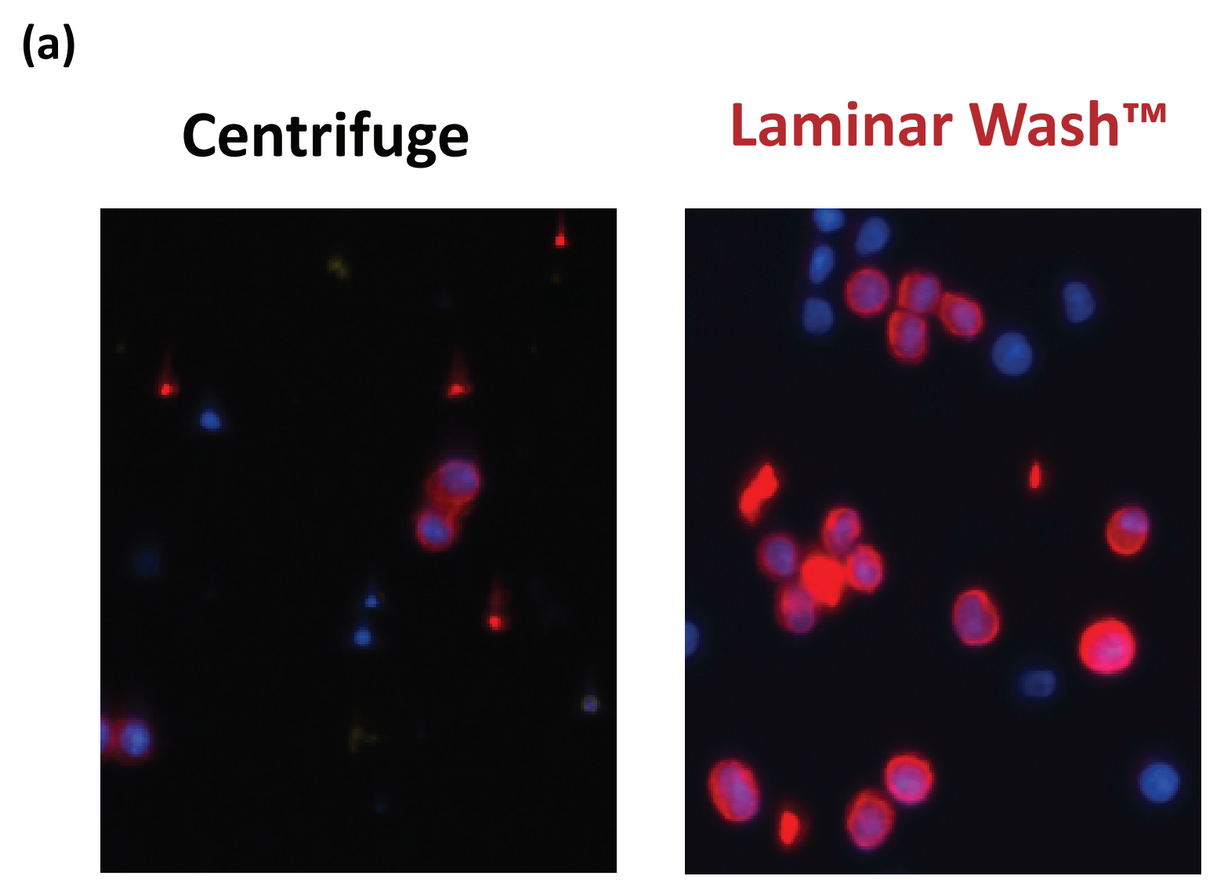
(a)
Centrifuge
Laminar Wash™

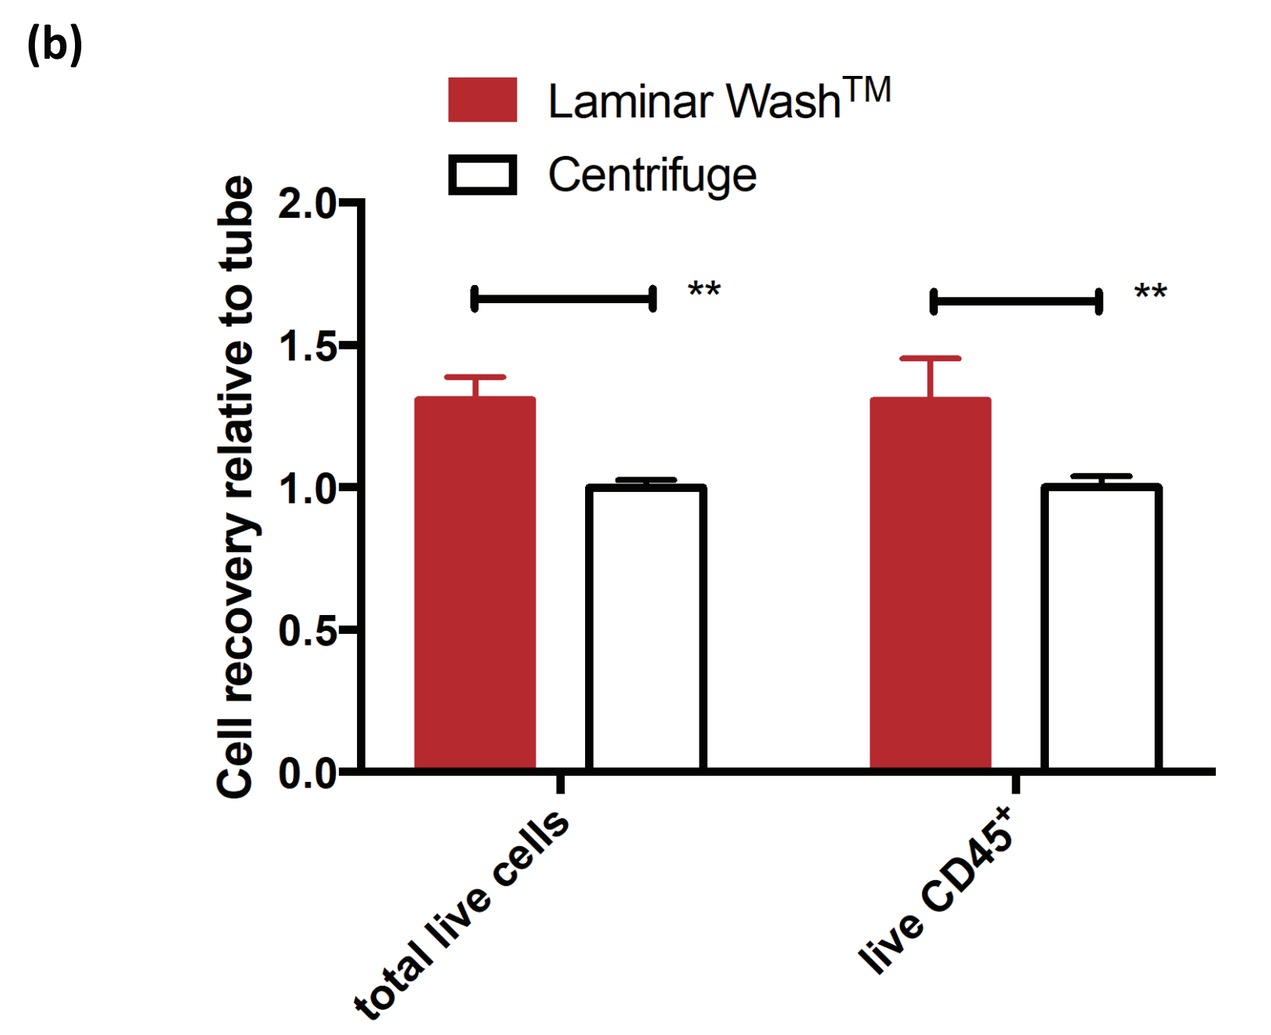
(b)
Laminar Wash™
Centrifuge
Cell recovery relative to tube
2.0
1.5
1.0
0.5
0.0
**
total live cells
live CD45+

Fig 3(a) In processing of low cell numbers (50-100), LW retained higher cell count with better cellular morphology and brighter endogenous fluorescent expression. The dim protein expression is evidence of mechanic stresses caused by centrifugation.
Data from a biotech company in San Diego

Fig 3(b) In subcutaneous murine melanoma tumor samples, LW retained 40% more live cells than centrifuge, with less tumor debris and better antibody staining.

LAMINAR WASH™ INCREASES % LIVE CELLs for TILs and LEADS TO HIGHER hCD45+ POPULATION

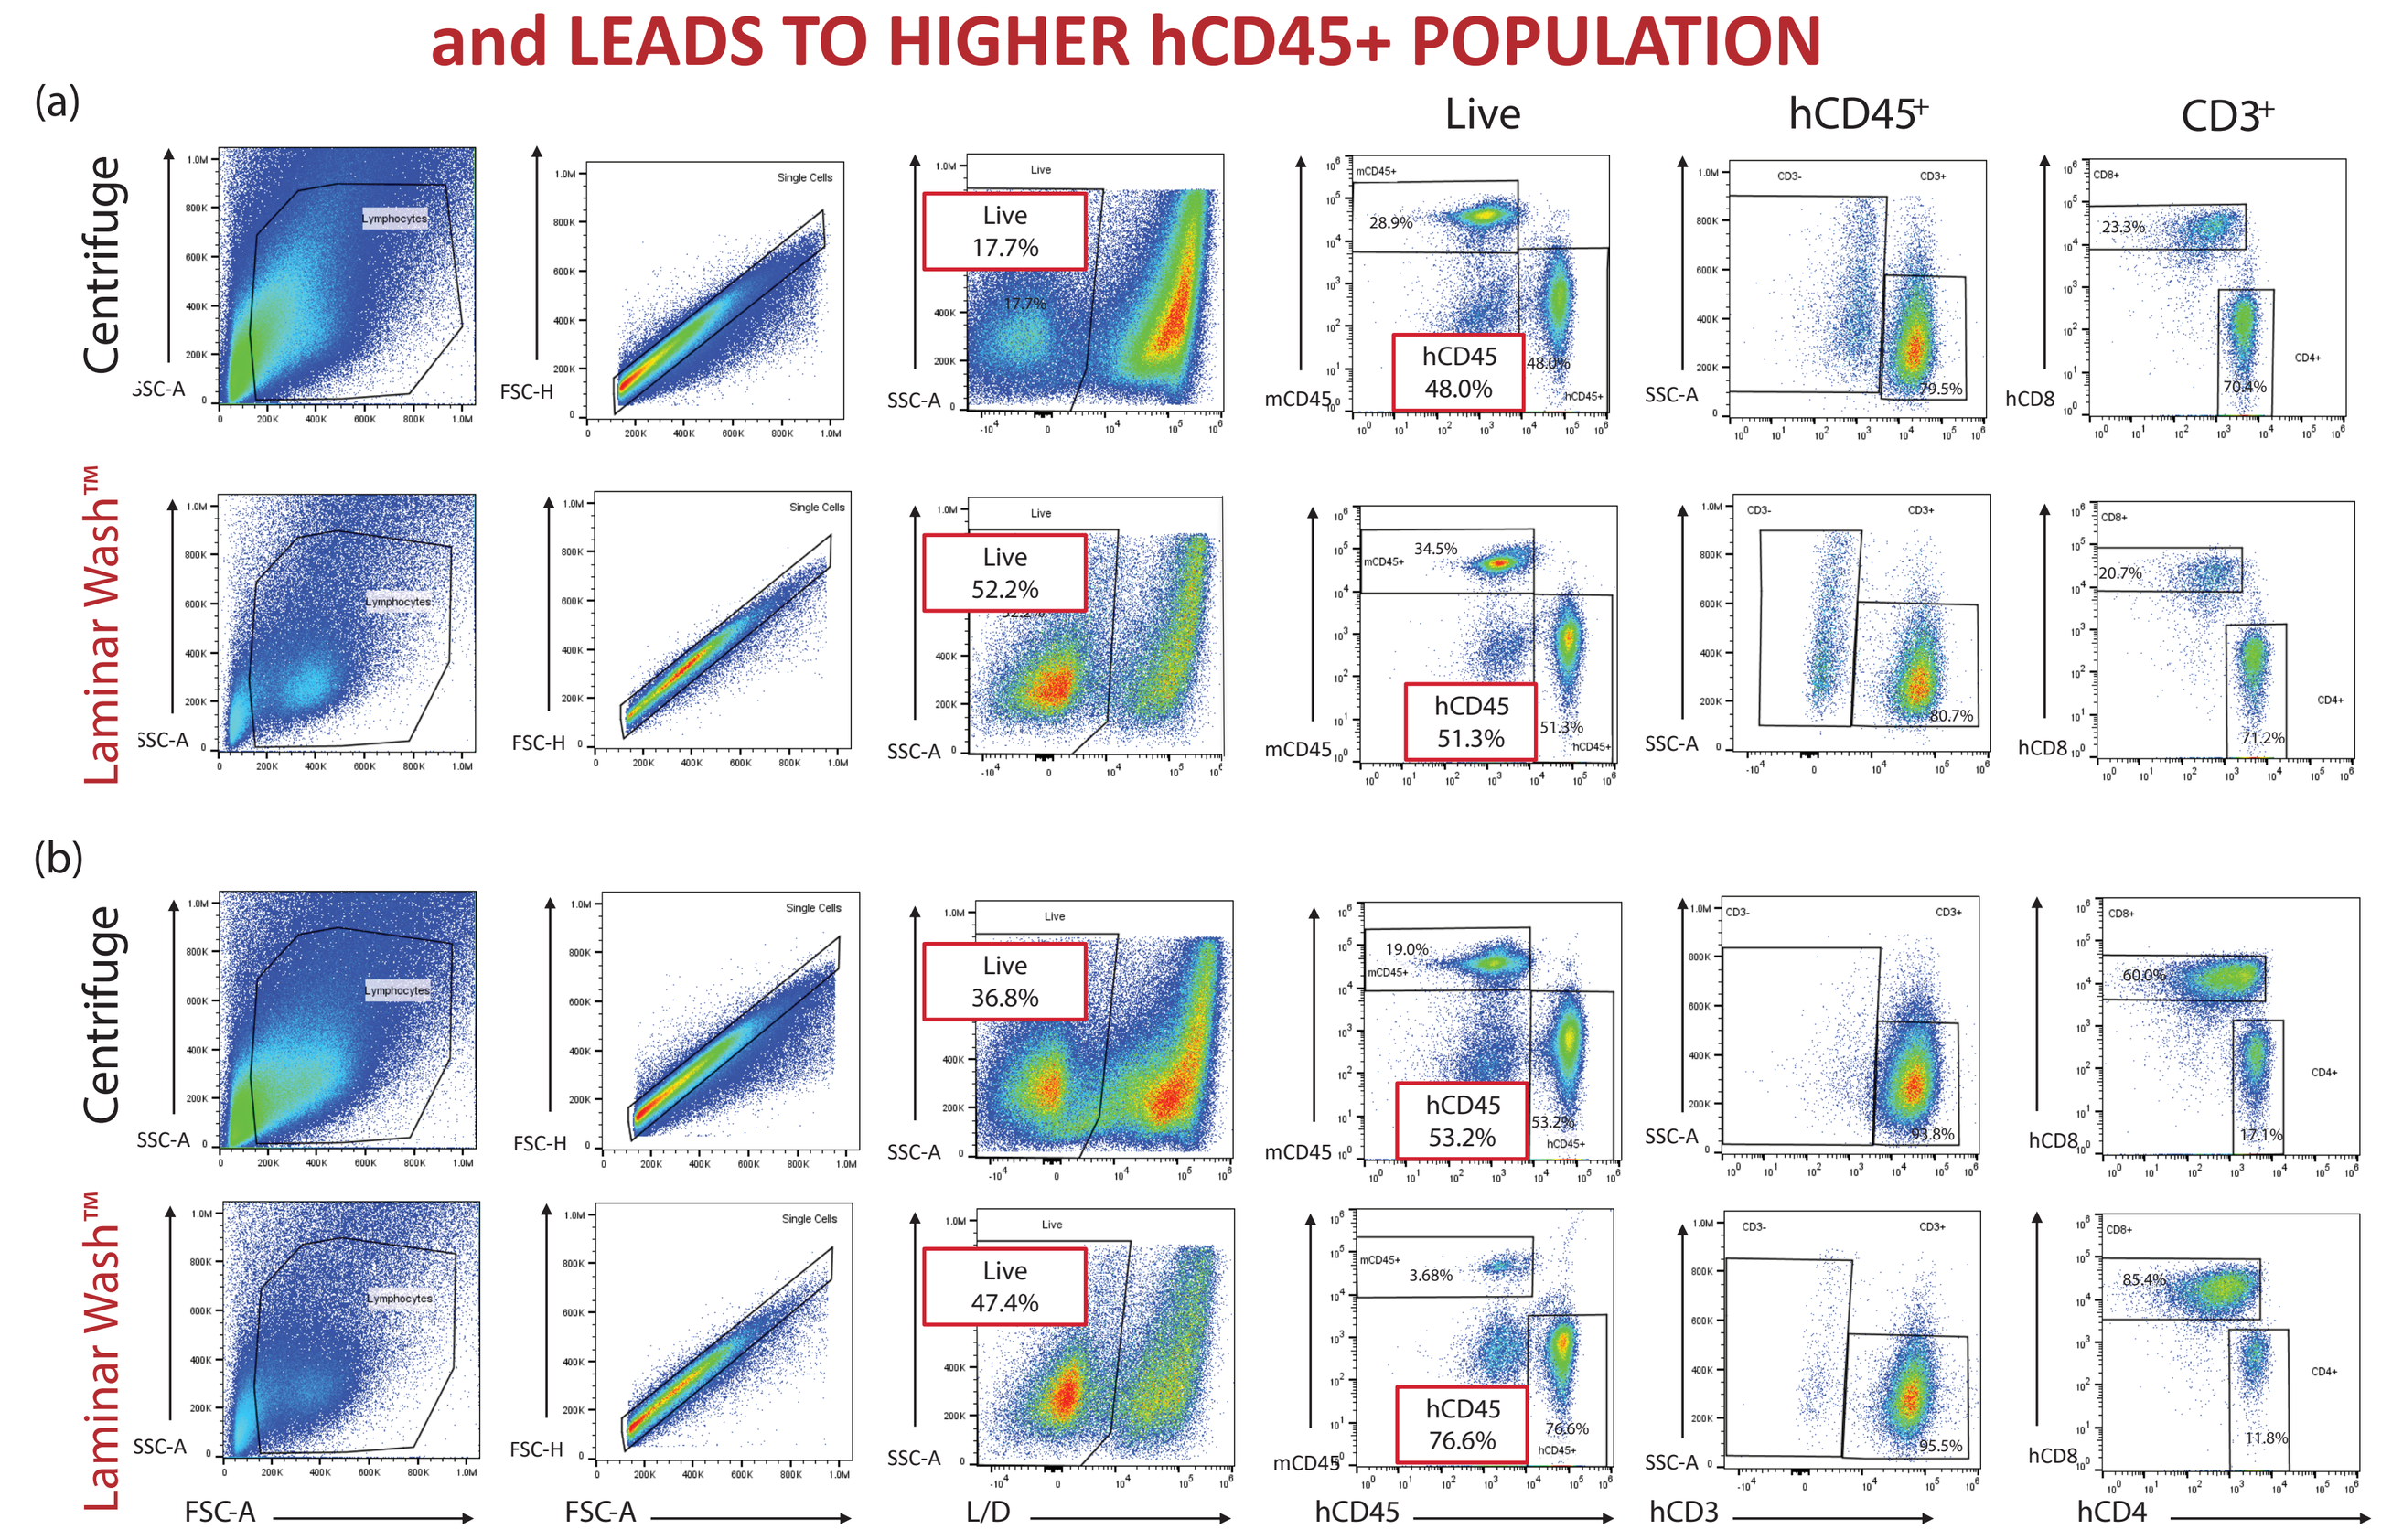
(a)
(b)
Live
hCD45+
CD3+
Centrifuge
Laminar Wash™
Live 17.7%
hCD45 48.0%
Live 52.2%
hCD45 51.3%
Live 36.8%
hCD45 53.2%
Live 47.4%
hCD45 76.6%
FSC-A
L/D
hCD45
hCD3
hCD4

Fig 4 A549-inoculated humanized mouse **(a)** unchallenged tumors **(b)** challenged tumors

LW™ LEADS TO MORE EFFECTIVE INTRACELLULAR STAINING of RARE IMMUNE CELL POPULATIONS

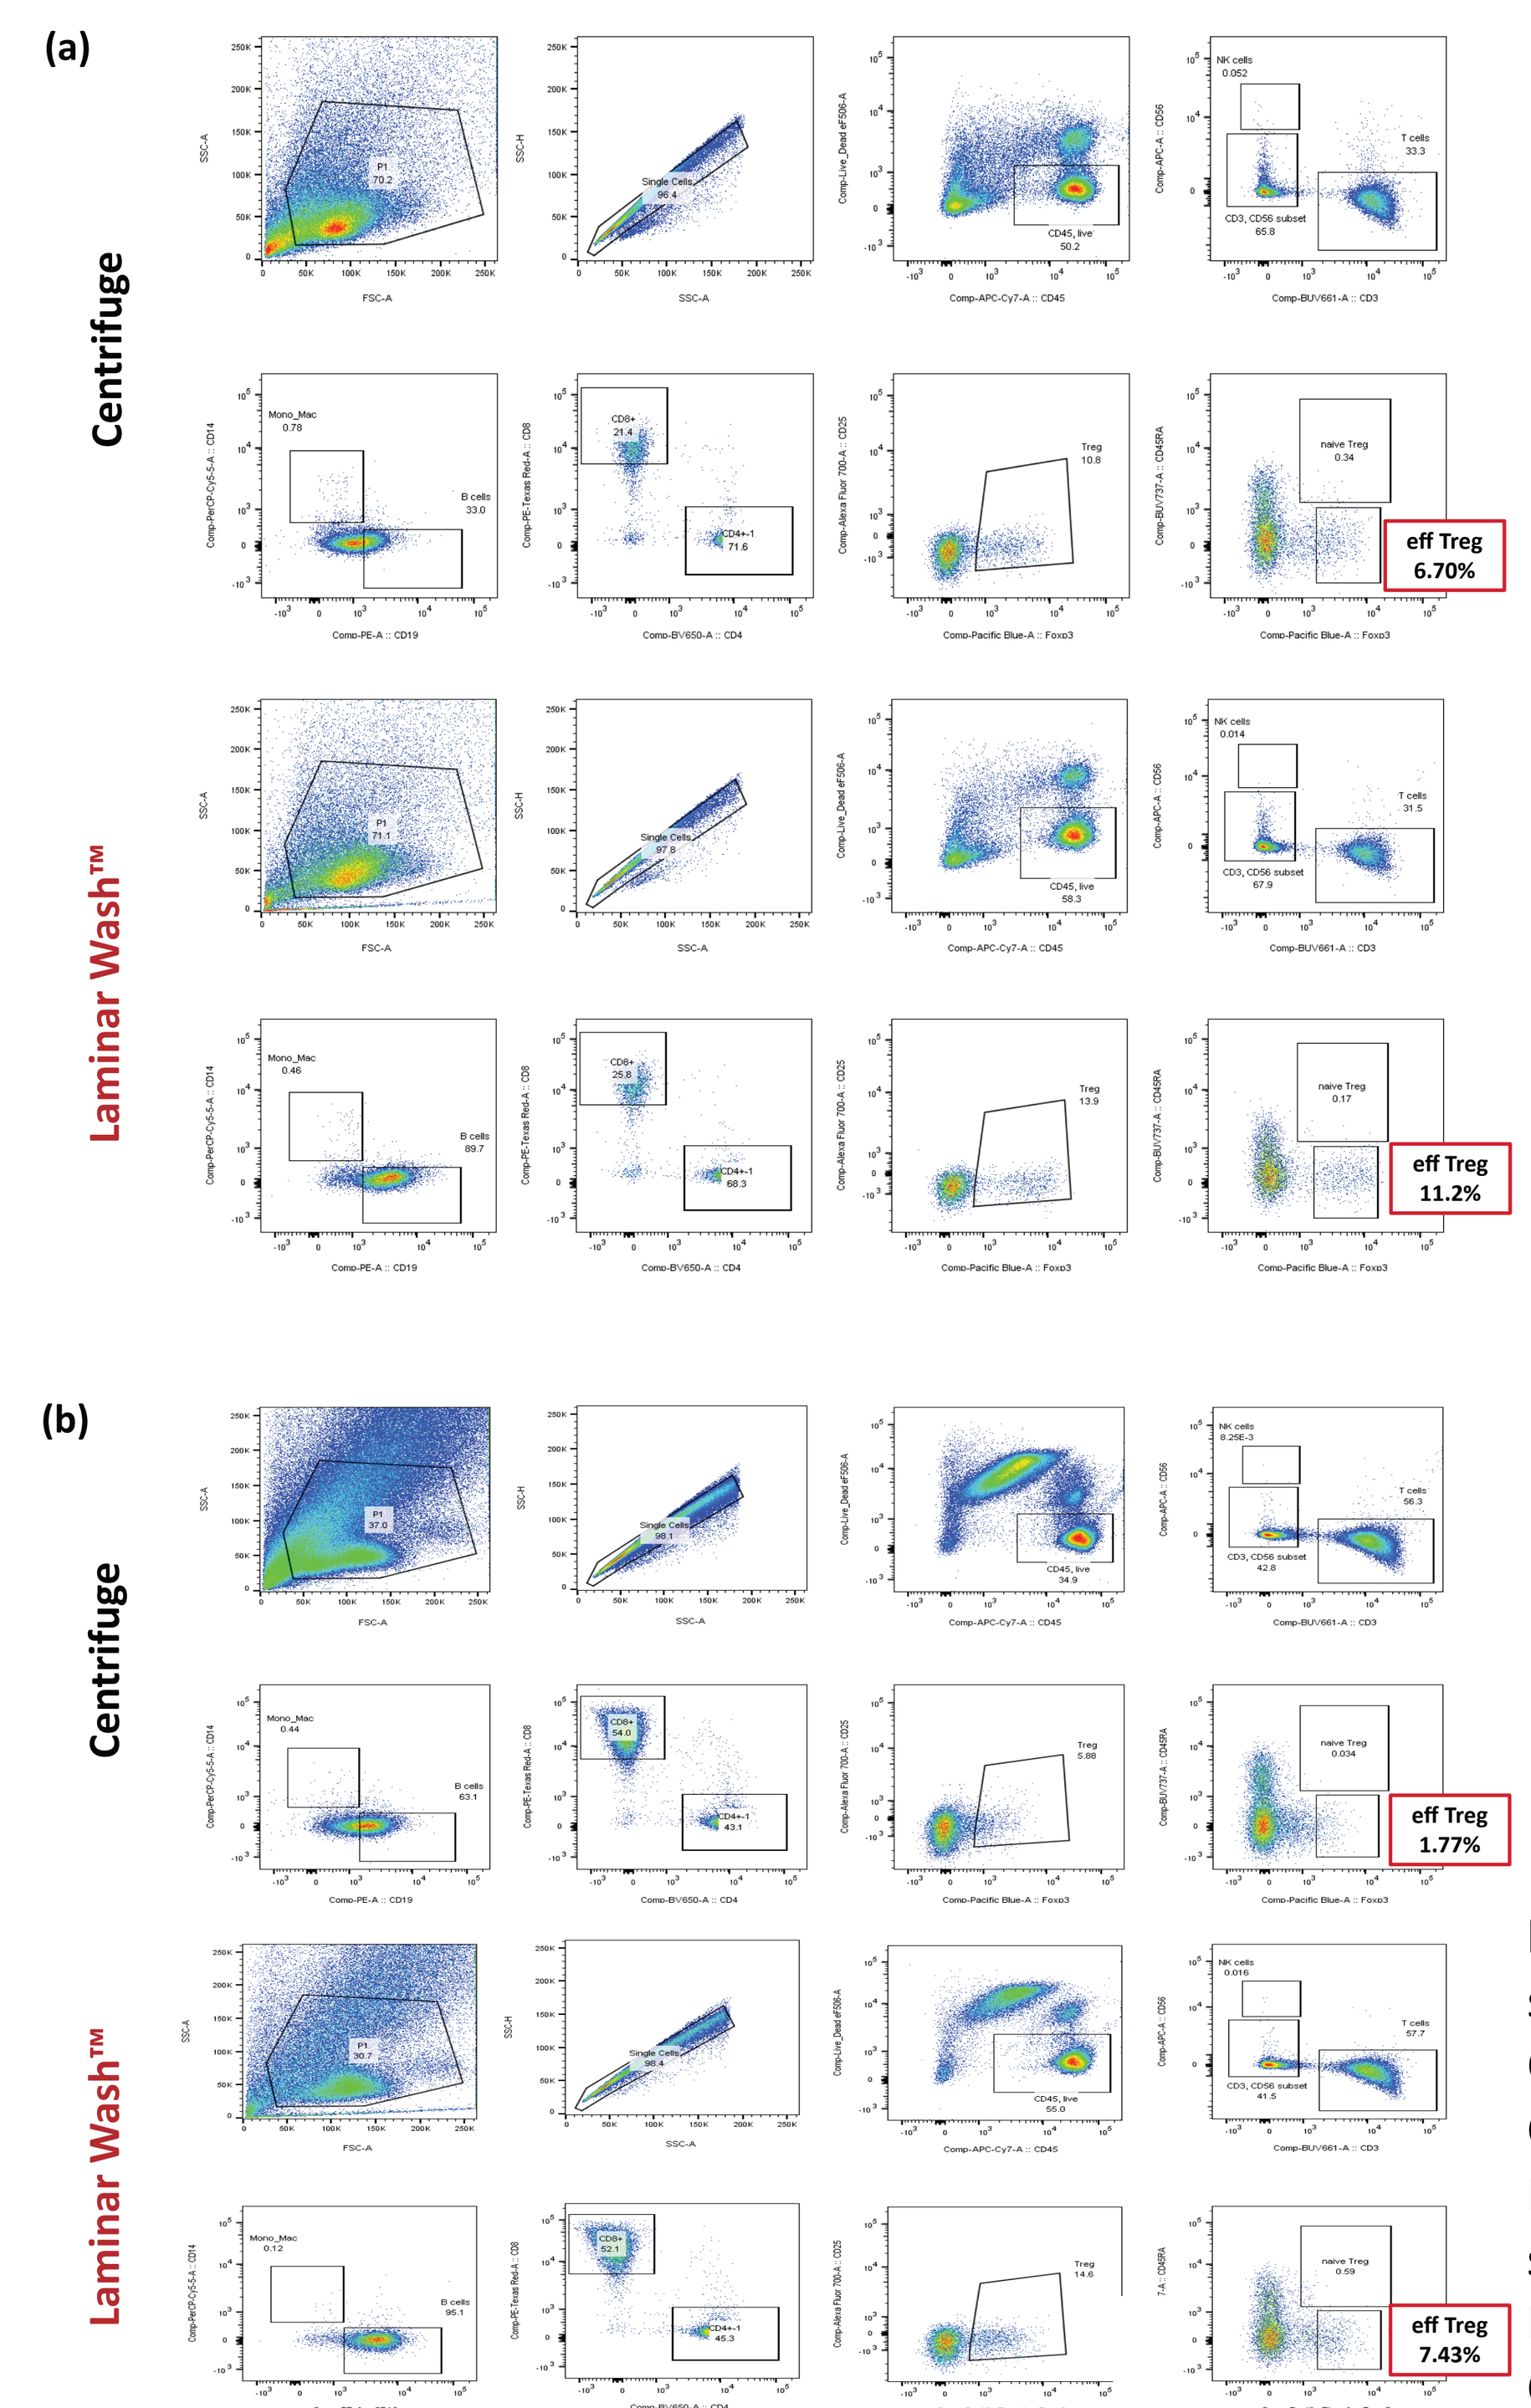
(a)
(b)
Centrifuge
Laminar Wash™
eff Treg 6.70%
eff Treg 11.2%
eff Treg 1.77%
eff Treg 7.43%

Fig 5 Immunophenotyping of various immune cell subsets on cryopreserved melanoma samples **(a) SK-MEL-919** and **(b) SK-MEL-954** by centrifugation and Laminar Wash methods

Fig 5(c) Percentages of CD4+ subsets: CD25+Foxp3+, CD45RA-Foxp3hi, and CD45RA+Foxp3+ cells in PBMCs and melanoma samples processed by Laminar Wash (black) vs. Centrifugation (grey) methods

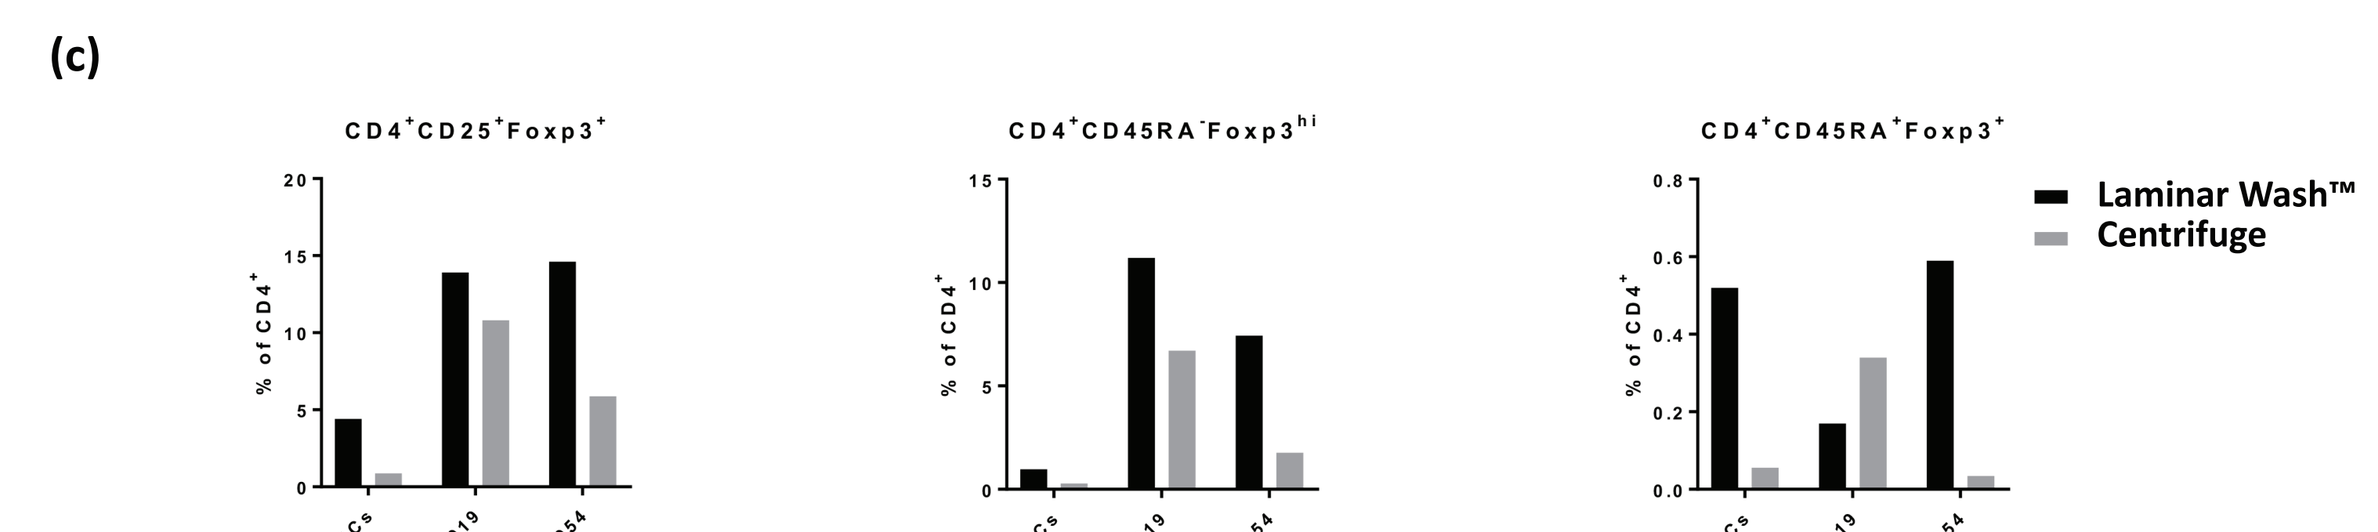
(c)
CD4+CD25+Foxp3+
CD4+CD45RA-Foxp3hi
CD4+CD45RA+Foxp3+
% of CD4+
PBMCs
SK-MEL 919
SK-MEL 954
Laminar Wash™
Centrifuge

LAMINAR WASH™ ENABLES ACCURATE IDENTIFICATION of TUMOR INFILTRATING LYMPHOCYTE (TIL) POPULATION

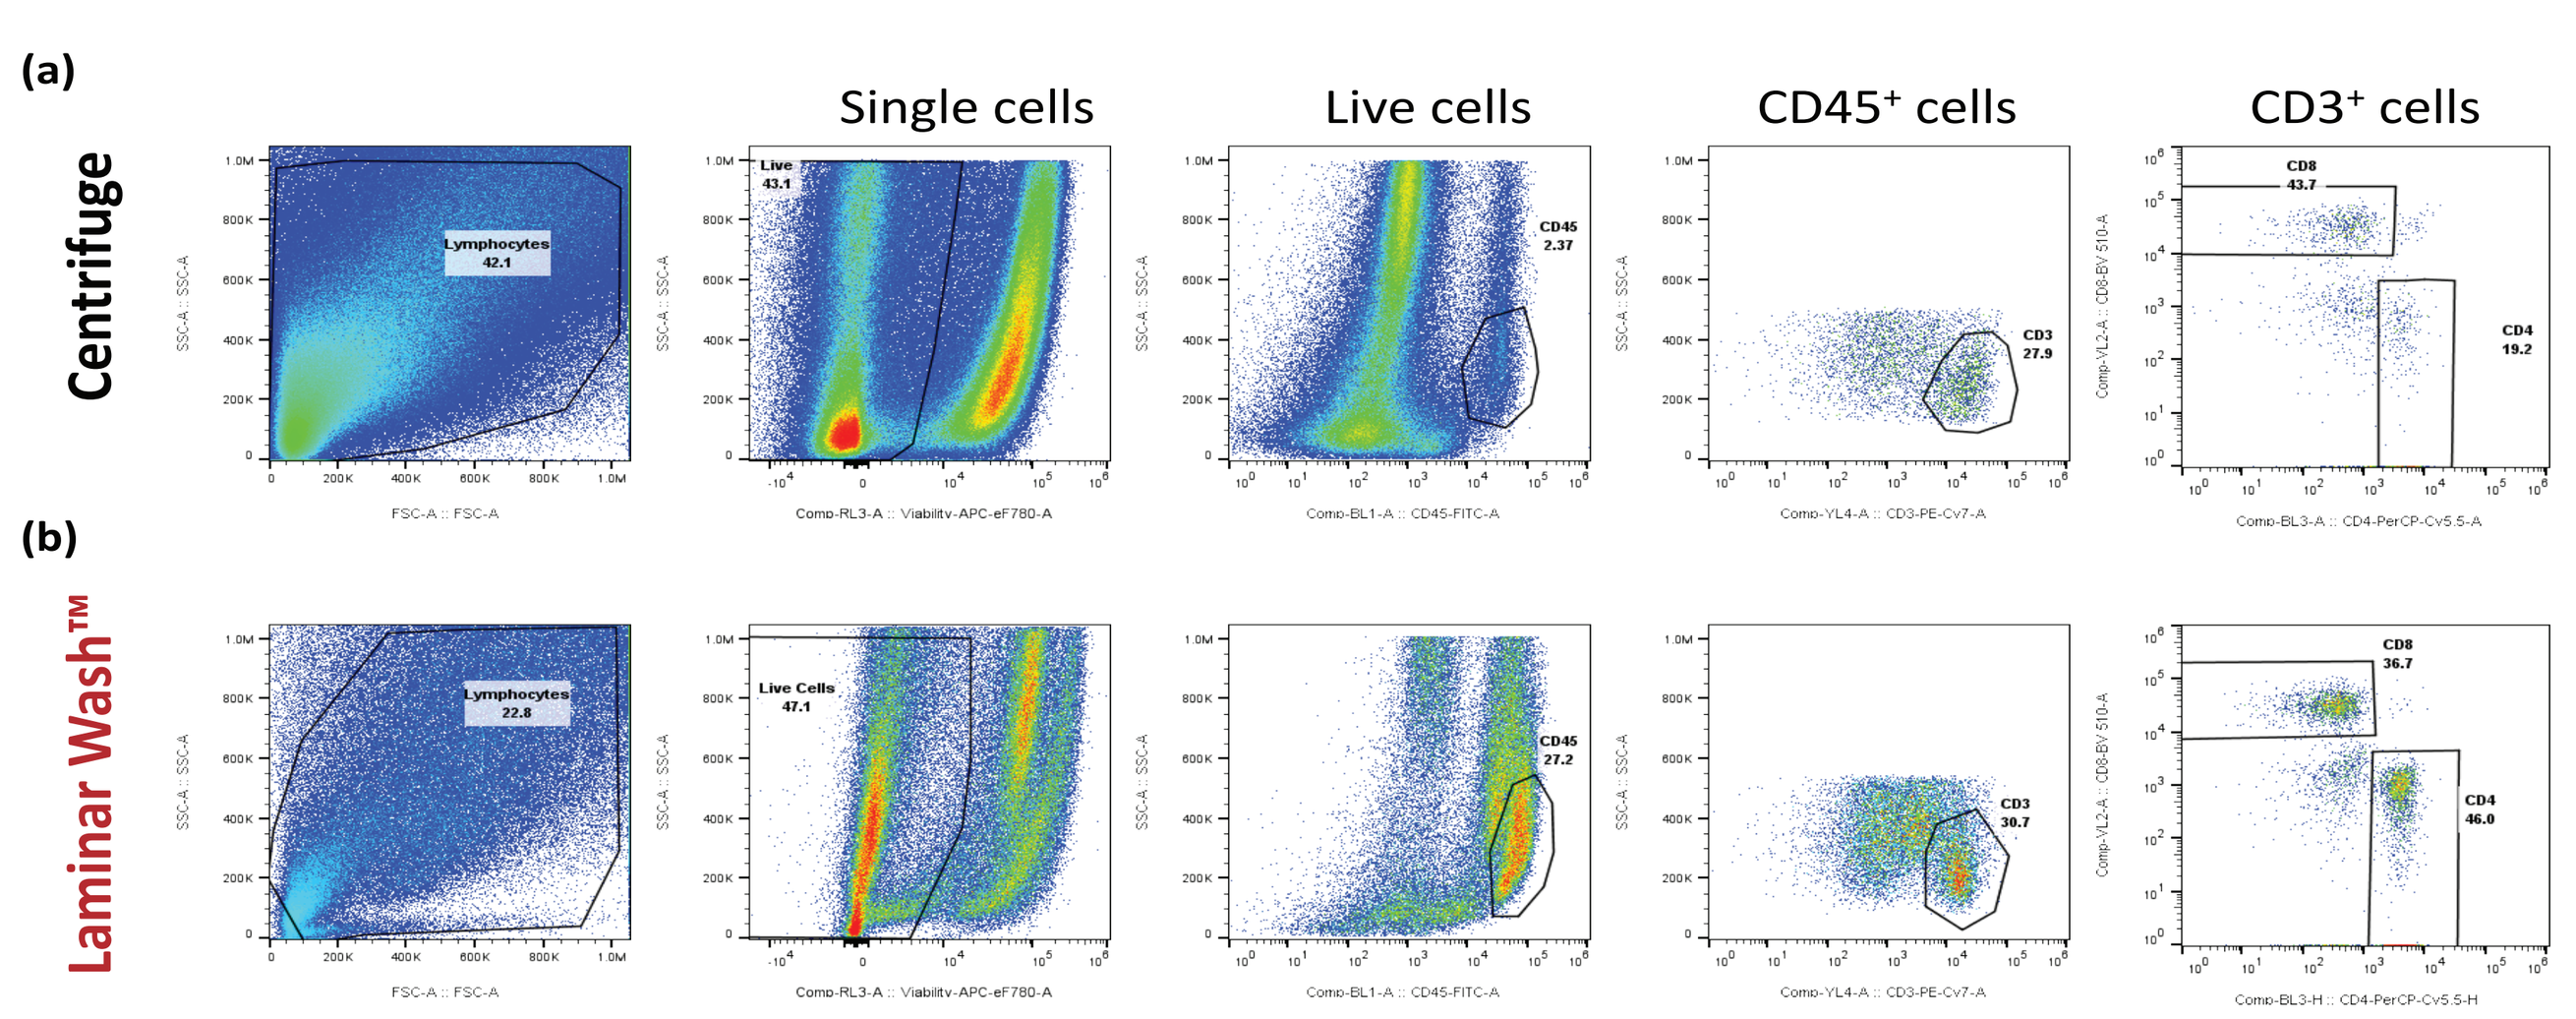
(a)
(b)
Centrifuge
Laminar Wash™
Single cells
Live cells
CD45+ cells
CD3+ cells

Fig 7(a) Centrifuge wash method – cell loss and mechanical stress through sequential pelleting and resuspension **Fig 7(b) Laminar Wash™** method – less tumor debris, but higher retention of TILs and better resolution of populations

LAMINAR WASH™ LEADS to IMPROVED CELL VIABILITY of TILs and LESS CYTOMETER CLOGGING

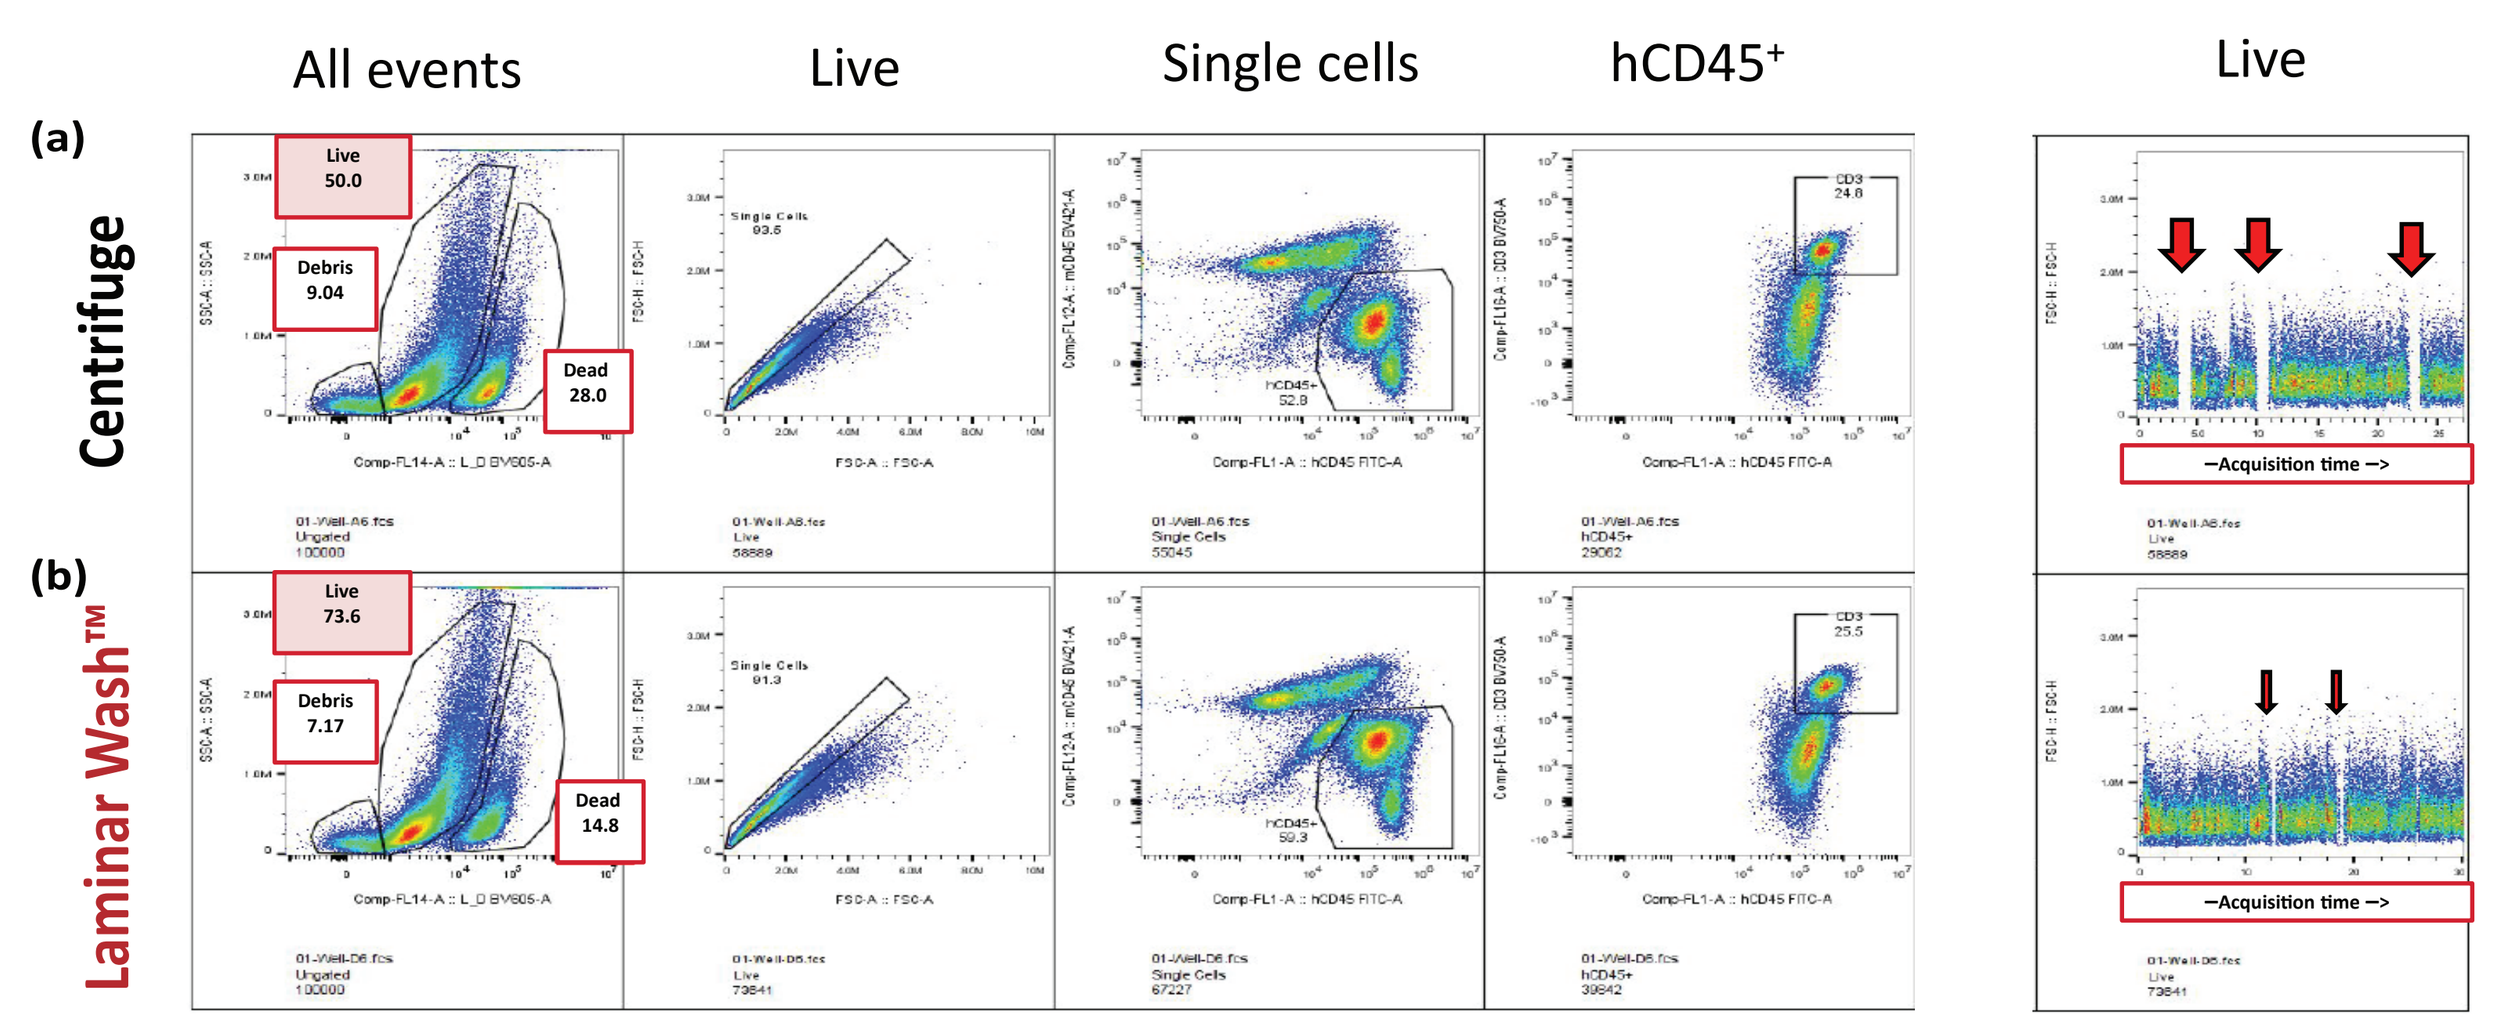
All events
Live
Single cells
hCD45+
Live
(a)
(b)
Centrifuge
Laminar Wash™
Live 50.0
Debris 9.04
Dead 28.0
Live 73.6
Debris 7.17
Dead 14.8
—Acquisition time —>

Fig 8 FACS plots by **(a) centrifuge** vs. **(b) Laminar Wash™** methods. The dissociated tumor samples from Raji-inoculated Balb/c mice showed higher viability and increased debris removal with Laminar Wash™, leading to less clogging and more consistent reading.

CONCLUSIONS

-

Laminar Wash™

1. produces consistent data with low coefficient of variance through automation
2. improves retention and better retains endogenous protein expression
3. increases % live cells for TILs and leads to higher hCD45+ population
4. more effectively removes signal background leading to true positive signal detections
5. enables accurate identification of TILs
6. leads to more effective intracellular staining of rare immune cell populations

ACKNOWLEDGEMENTS

We would like to give special thanks to the following scientists: Dr. Arnaud Colantonio at Adicet Bio (Fig 2), Dr. Christoph Eberle at Charles River Laboratories in Worcester, MA (Figs 4&7), Drs. Roberta Zappasodi, Isabell Schulze and Taha Merghoub (Figs 5&6) and Dr. Thomas Tan, Jonathan Hsu, and Benjamin Keller at Elstar Therapeutics, Cambridge, MA (Fig 8) for sharing their valuable insights and data.

Curiox Biosystems